Treatment and Economic Challenges when Treating Patients with Agitation Associated with Schizophrenia or Bipolar Disorder in the Emergency Department

Mae Kwong, PharmD[1] and Sonja Hokett, PharmD, MS, MSc[1]
[1]BioXcel Therapeutics, Inc., New Haven CT

OBJECTIVE

Episodes of acute agitation associated with schizophrenia (SCZ) or bipolar disorder (BPD) pose treatment and economic challenges for patients and staff.

Occurrence of staff injuries, physical restraint use, extended inpatient admissions, and increased emergency department (ED) boarding create a complex clinical environment.

Identifying and quantifying the impact of these treatment challenges and economic drivers is critical to effective patient management strategies.

METHODS

- Two separate analyses of medical and pharmacy claims data [Clarivate Real World Data between 9/2015 and 4/2022 and Healthcare Cost and Utilization Project (HCUP) Nationwide Emergency Department Sample (NEDS) from 2018] quantified prevalence and direct medical costs from ED and inpatient psychiatry visits.
- These data were combined with a multicenter retrospective chart review and published data to provide a comprehensive overview of the clinical and economic burden of acute agitation episodes in patients with SCZ and BPD.

CONCLUSION

- Treatment of agitation associated with SCZ and BPD is challenging and results in higher healthcare utilization and higher patient mortality risk.
- Direct medical costs are increased from ED and inpatient admission incidence rates and length of stays, increased patient physical restraint use, increased staff injuries, and ED boarding.
- Detailing and quantifying these clinical and economic patient treatment challenges can assist in evaluating the current therapeutic paradigms and selecting effective and safe treatment options which may improve patient outcomes and lower staff burden.

High Utilizers of Health Resources for Agitation[1]

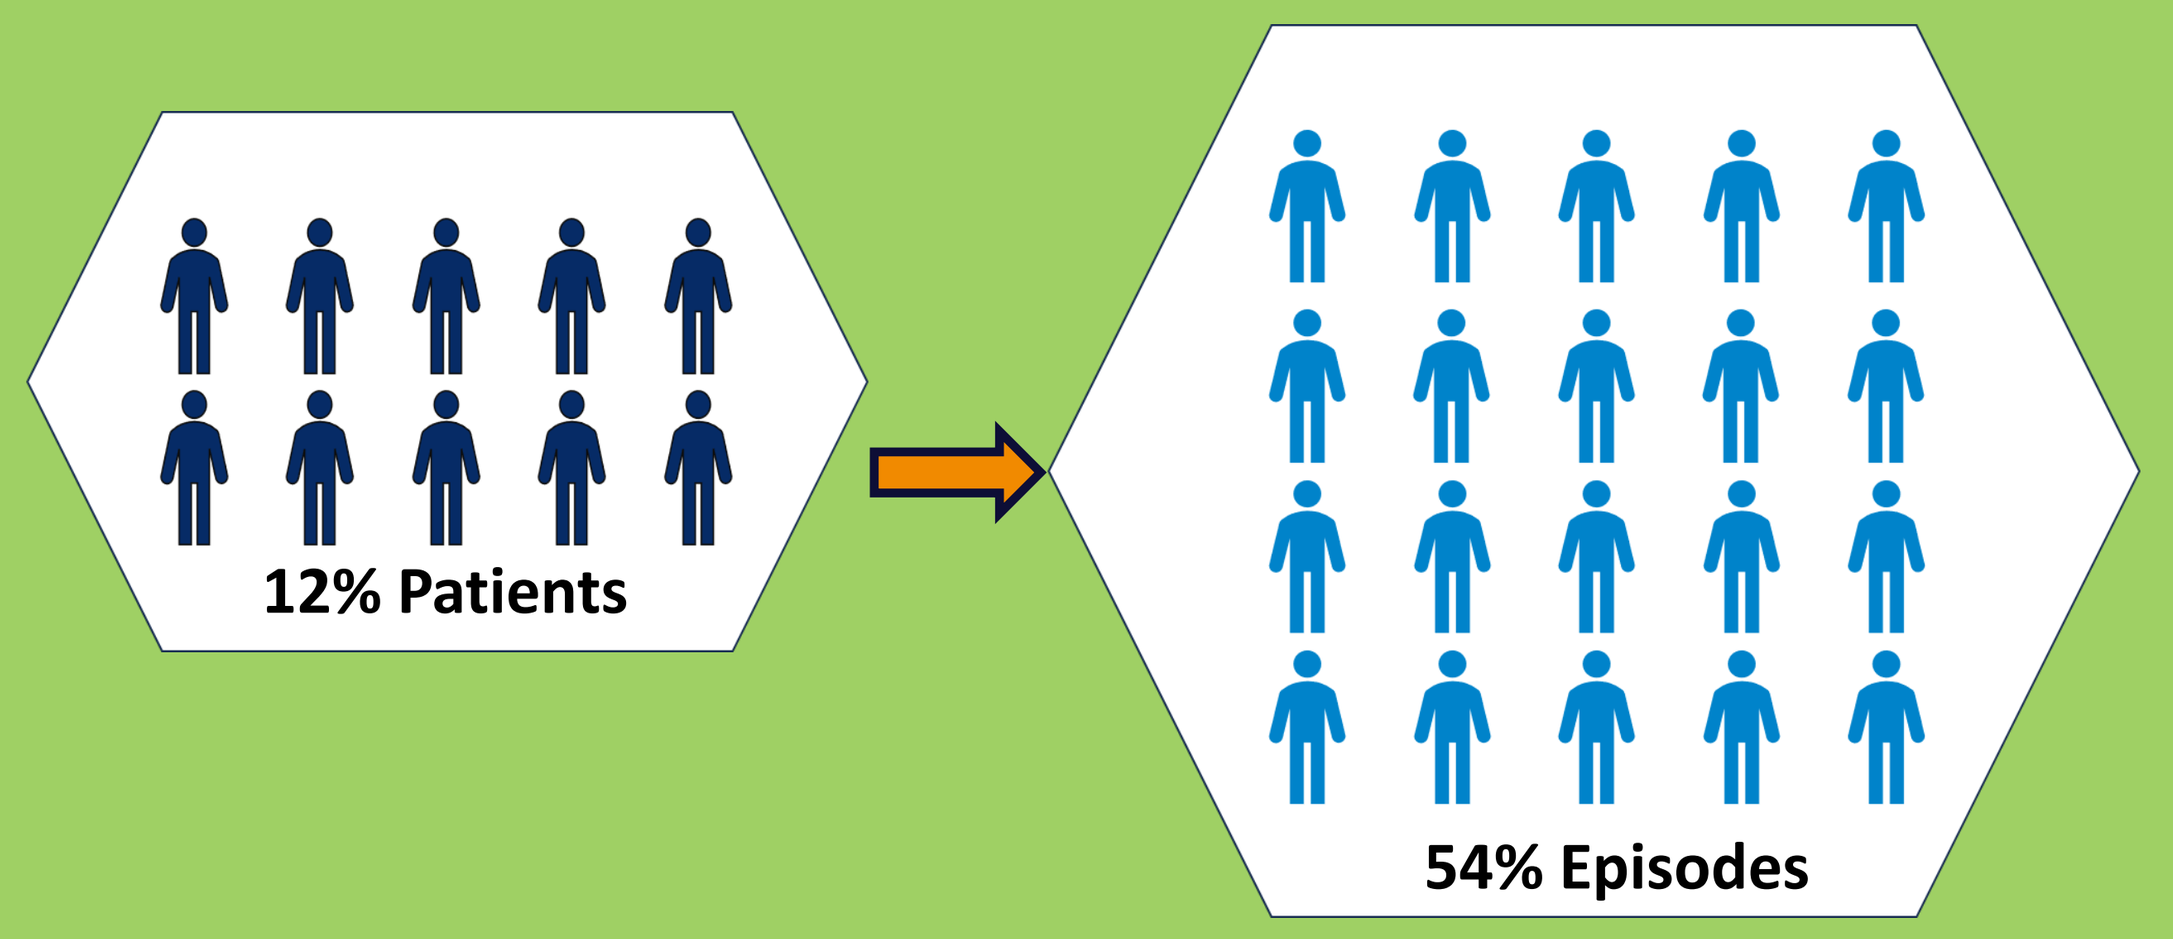
12% Patients
54% Episodes

- **12% of patients account for 54% of agitation episodes**

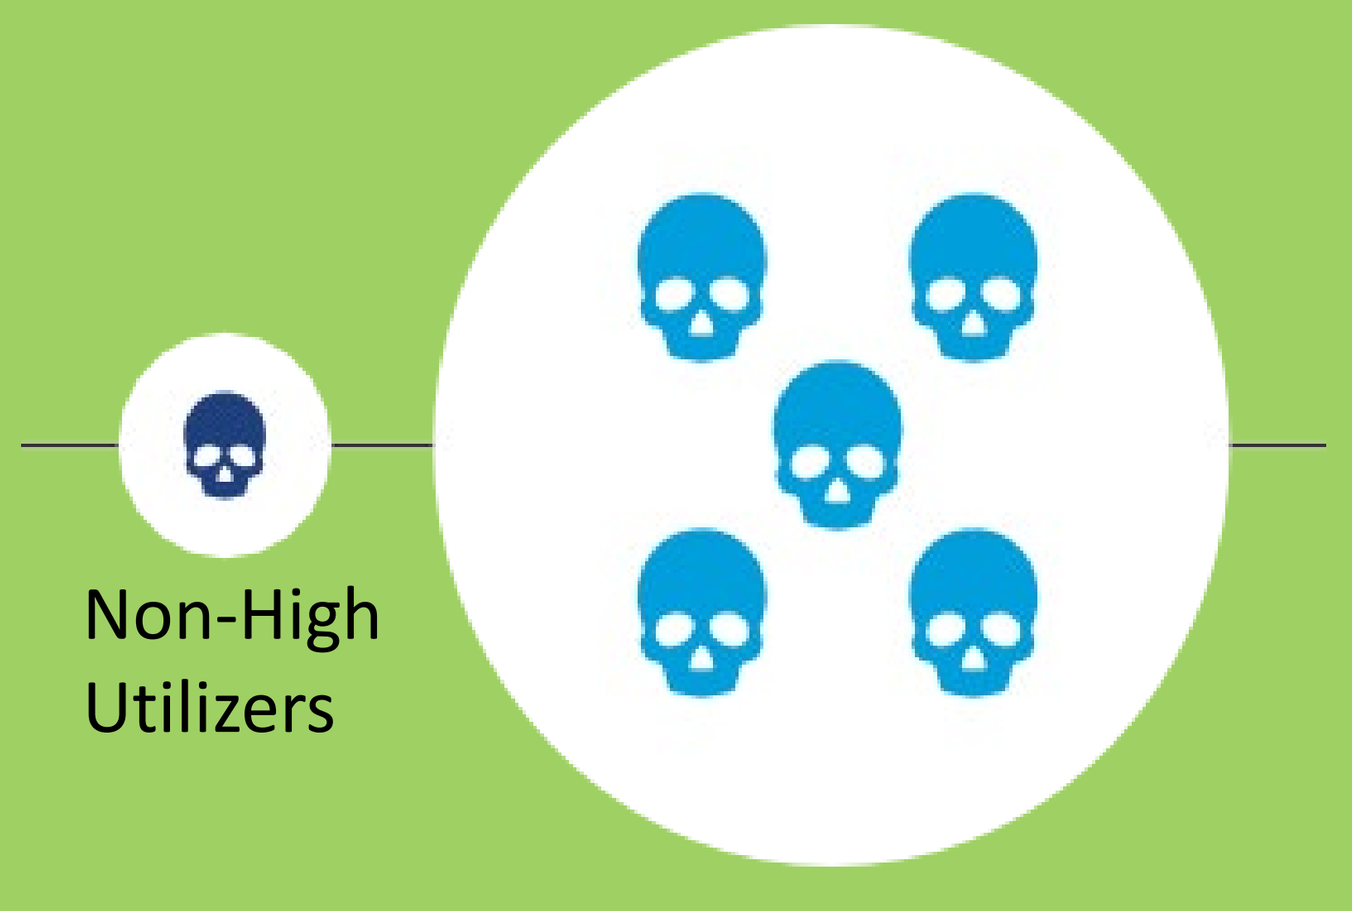
Non-High Utilizers
High Utilizers

- **5 times higher mortality risk**

Hospital Revisits

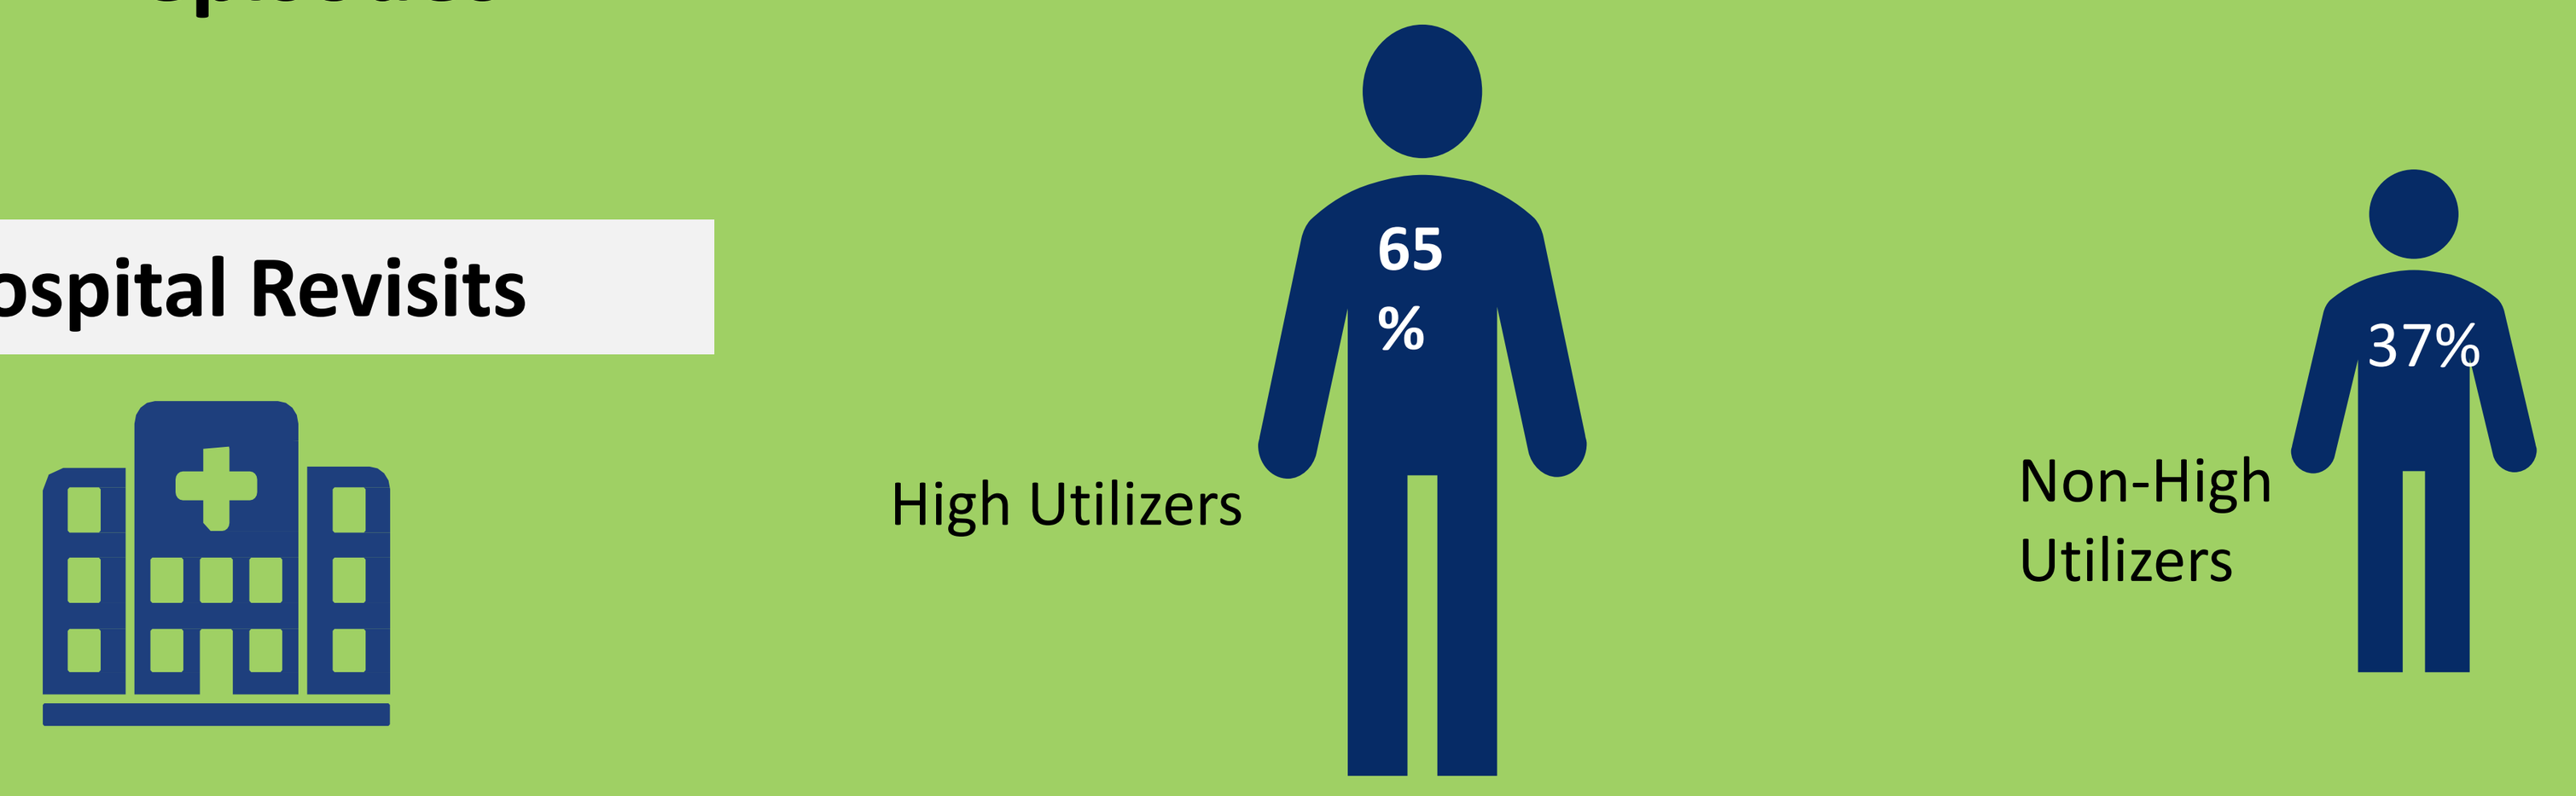
65 %
High Utilizers
37%
Non-High Utilizers

- **Nearly double 30-day readmission rate (65% vs 37%)**

Costs and Health Resource Utilization Impact of Agitation

- **INPATIENT ADMISSIONS[2,3]:**
 $11,000 (SCZ) & $7,600 (BPD)
- **27% PHYSICALLY RESTRAINED[4]:**
 $1,511 (SCZ) and $1,233 (BPD)[2,3]
 - **4.2 additional hours in ED**
 - **8% higher inpatient admission rate**
 - **1.45 days longer inpatient stay**
 - **22% staff injury incidence**
- **ED BOARDING[5]:**
 $3,400 denying care of 2.2 patients
- **STAFF INJURY/WORKMAN'S COMPENSATION CLAIMS[6]:**
 $15,860 average claim (between 2006 and 2011)

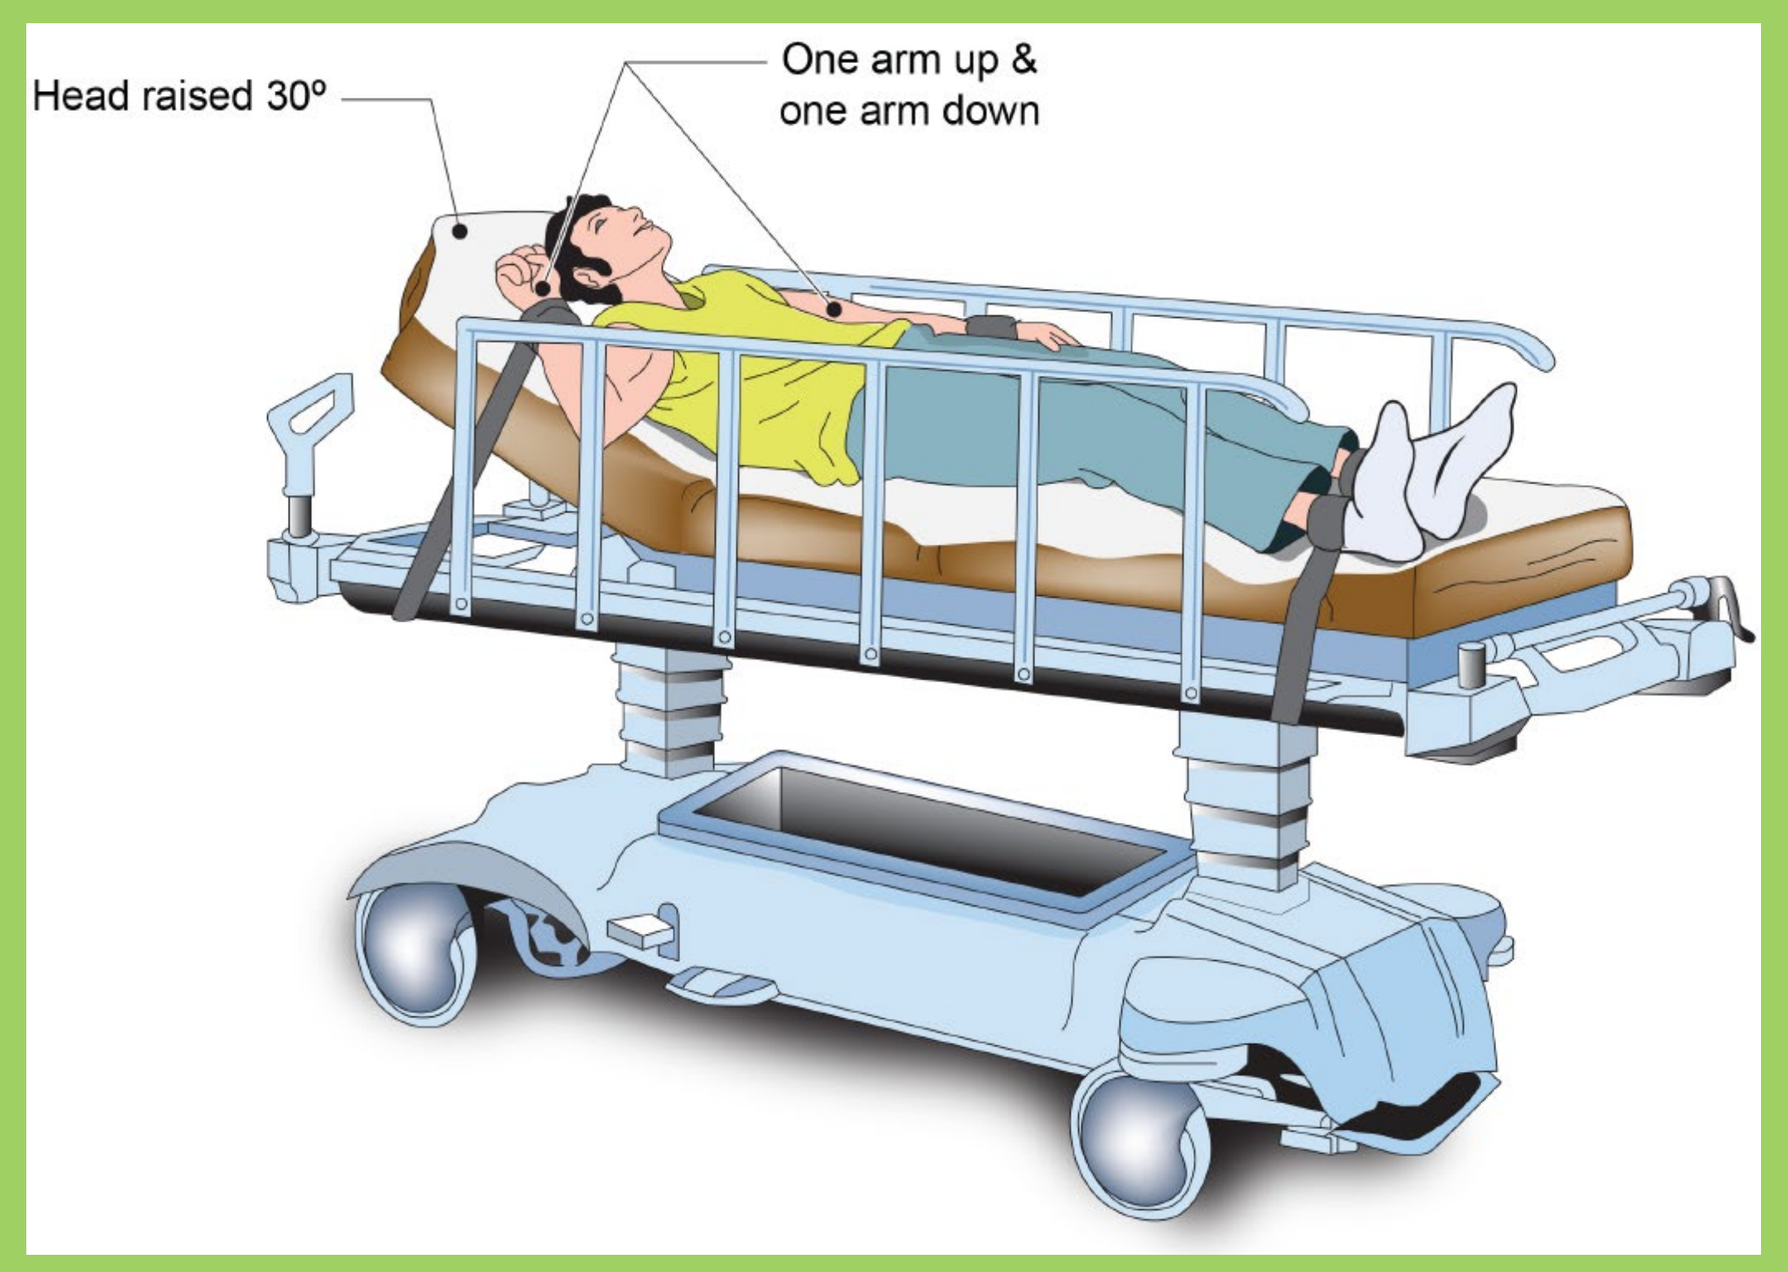
Head raised 30º
One arm up & one arm down

RESULTS

- **Within this population, high utilizers of healthcare resources comprise 12% of patients while accounting for 54% of agitation episodes. This subset of high-utilizing patients had 5 times the mortality risk and nearly double the 30-day hospital readmission rate (65% vs 37%) versus the lower-utilizing group.**
- **When treated in the ED, 27% of patients with agitation associated with SCZ or BPD were physically restrained contributing to additional direct medical costs of $1,511 (SCZ) and $1,233 (BPD). Components of these additional costs of physically restraining patients in the ED included 4.2 additional hours in the ED, 8% higher inpatient admission rate, 1.45 days longer inpatient stays, and 22% staff injury incidence.**
- **Total inpatient admission costs were $11,000 (SCZ) and $7,600 (BPD). Hospital staff injury Workman's Compensation claims were an average of $15,860 (between 2006 and 2011). Additional costs for ED patient boarding are $3,400 denying care of 2.2 patients.**
- **High resource utilizer patients with SCZ and BPD should be identified for enhanced treatment strategies to reduce economic burden to patients, staff, and health systems.**

References: 1. Kwong M, et al. Poster presented at AMCP 2023. March 21-24, 2023, San Antonio, TX. 2. Hokett S & Kwong M. Poster presented at ISPOR 2022. May 15-18, 2022, Washington, DC. 3. Hokett S & Kwong M. Poster presented at ISBD 2023. June 22-25, 2023, Chicago, IL. 4. Data on File. BioXcel Therapeutics, Inc. 5. Nicks BA, et al. *Emer Med Intl.* 2012(5):360308. 6. Occupational Safety and Health Administration. Accessed August 1, 2023. https://www.osha.gov/sites/default/files/1.2_Factbook_508.pdf

Presented at the Academy of Managed Care Pharmacy Annual Meeting, April 15-18, 2024, New Orleans, LA